F2-356

Selection of Optimal Antisense Clones of *Bacillus anthracis* for Drug Discovery Efforts

V. BROWN-DRIVER, K. GC, D. REYES, M. STIDHAM, J. FINN, R. HASELBECK, M. HILGERS, K. J. SHAW;
Trius Therapeutics, San Diego, CA.

Vickie Driver
Trius Therapeutics
6310 Nancy Ridge Dr. Ste. 105
San Diego, CA 92121
858-452-0370
vdriver@triusrx.com

Abstract

Background: Antisense induction selectively attenuates gene expression in bacteria. We reported previously that *B. anthracis* antisense clones are useful in determining mechanism of action and identifying essential genes (ICAAC 2005-F2075 and 2006-F21170 posters). Here we demonstrate the importance of examining multiple antisense clones for an individual target to identify optimally functional clones for drug discovery.
Methods: Several antisense clones for both *B. anthracis murA1* and *metRS1* were identified using our targeted single gene antisense screening approach. PCR products of *murA1* and *metRS1* were amplified, fragmented and cloned into a xylose-inducible vector. The antisense library was transformed into an avirulent strain of *B. anthracis* and screened for xylose sensitivity. Xylose sensitive clones were tested for hypersensitivity to target-specific compounds.
Results: Unique antisense clones showed different degrees of hypersensitivity to compounds that specifically inhibit the target. Of 12 *murA1* antisense clones only 6 showed >5-fold hypersensitization to the MurA specific antibiotic fosfomycin upon xylose induction. Similarly, only 4 of 6 *metRS1* antisense clones showed >5-fold hypersensitization to the MetRS1 specific compound Rx-100019. When a panel of antibiotics was tested against two *murA1* antisense clones, only one clone was hypersensitive to a number of penicillins and cephalosporins.
Conclusions: All antisense clones do not behave the same way in hypersensitivity screens. Thus, clones should be carefully evaluated when optimizing antisense assays for drug discovery efforts.
This work was supported by NIH SBIR Grants R43 AI069666 and R44AI53009.

Materials and Methods

Plasmids and Strains: Nonpathogenic and plasmidless *Bacillus anthracis* strain, UM23C1-1 was used as the screening strain (11). *E. coli* strains: DH5α and INV110 (*dam⁻/ dcm⁻*) (Invitrogen) were used for library manipulation. Xylose inducible shuttle vector pBAX2 (1,2,3) was used as antisense expression vector.
Media and Growth Conditions: *E. coli* was grown in LB broth and agar plates and selected in the presence of carbenicillin at 100 µg/mL. *B. anthracis* strain UM23C1-1 was grown in Brain Heart Infusion (BHI) broth or agar plates with or without 10 µg/mL chloramphenicol.
Antisense Library Construction and Screening: Both *B. anthracis* genes *murA1* and *metRS1* were PCR amplified and fragmented by sonication. The random fragments were end polished by treating with T4 polymerase and Klenow fragement polymerase. This frament library was cloned into SmaI site of the pBAX2 cloning vector and transformed into DH5α cells. The library was passaged through INV110 and subsequently transformed into *B. anthracis* by electroporation. Clones showing growth inhibition in the presence of 2% xylose were selected for additional characterization, which included sequencing of the plasmid insert and determination of the concentration dependence of xylose growth inhibition.

Results

Figure 1: Multiple Antisense Clones Obtained for *murA1* gene Target

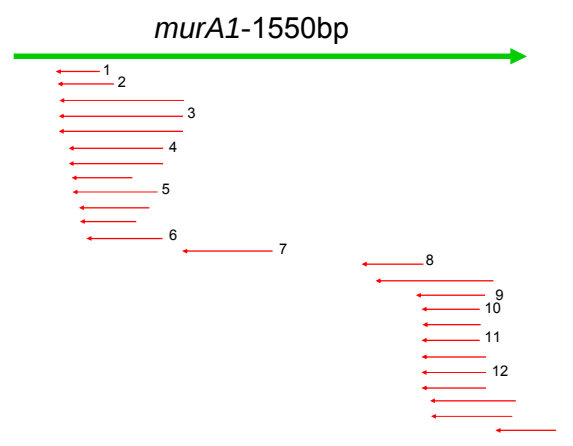
murA1-1550bp
1
2
3
4
5
6
7
8
9
10
11
12

- Multiple antisense clones for *murA1* were obtained in screening (←)
- Clones 1-12 were tested against phosphomycin

Figure 2: Specific Inhibition of Antisense

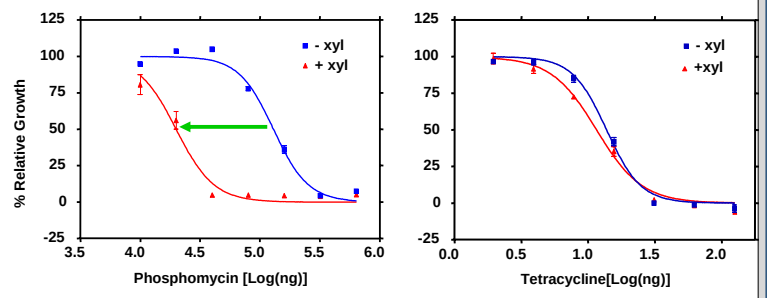
% Relative Growth
- xyl
+ xyl
Phosphomycin [Log(ng)]
- xyl
+xyl
Tetracycline[Log(ng)]

A. IC_{50} shift of *murA1* in the presence of phosphomycin

B. Tetracycline shows no shift

- Drug dose response curves were plotted in the presence (▲) or absence (■) of 40 mM xylose, and increasing concentrations of antibiotic
- No significant shift was observed for tetracycline
- An antisense-dependent 6.5-fold shift in IC_{50} was observed for phosphomycin

Figure 3: Differential Hyper-sensitization of *murA1* Antisense Clones by Phosphomycin

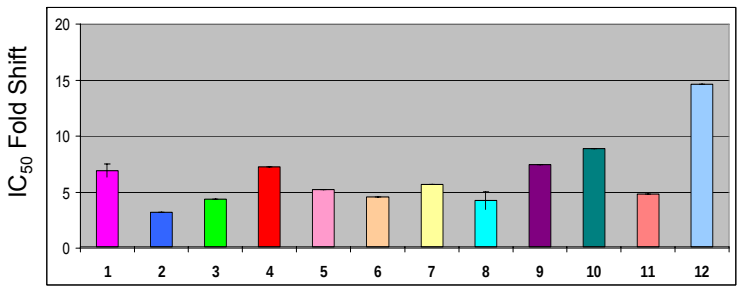
IC50 Fold Shift

murA1 antisense clones 1-12

- *murA1* Clones 1-12 were tested against phosphomycin showed differential hypersensitivity

Figure 4: Specificity Measurement of Drugs Against *murA1* Antisense Clones

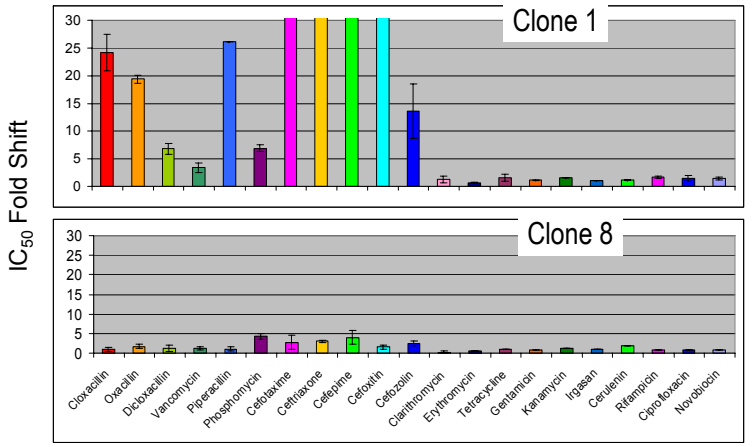
Clone 1
Clone 8
IC50 Fold Shift
Cloxacillin
Oxacillin
Dicloxacillin
Vancomycin
Piperacillin
Phosphomycin
Cefotaxime
Ceftriaxone
Cefepime
Cefoxitin
Cefozolin
Clarithromycin
Erythromycin
Tetracycline
Gentamicin
Kanamycin
Irgasan
Cerulenin
Rifampicin
Ciprofloxacin
Novobiocin

- Drug dose response curves were plotted in the presence and absence of inducer (xylose)
- The IC_{50} fold shift is calculated by dividing the uninduced IC_{50} by the induced IC_{50}

Figure 5: Multiple Antisense Clones Obtained for *metRS1* gene Target

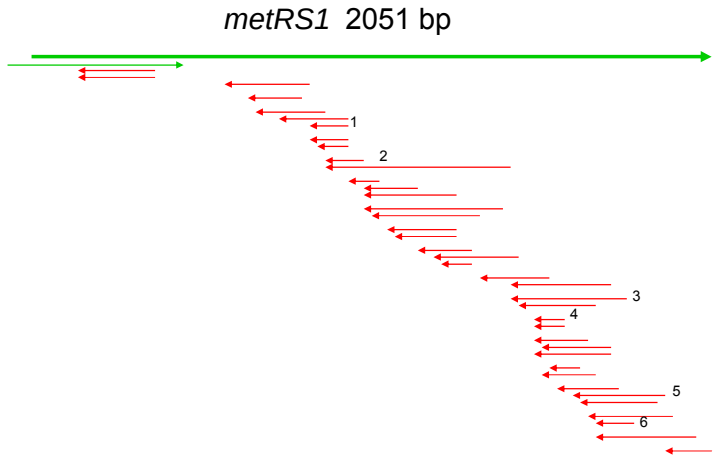
metRS1 2051 bp
1
2
3
4
5
6

- Multiple antisense clones for *metRS1* were obtained in screening (←)
- Clones 1-6 tested against RX-100019 which targets MetRS1

Figure 6: Differential Hyper-sensitization of *metRS1* Antisense Clones by RX-100019

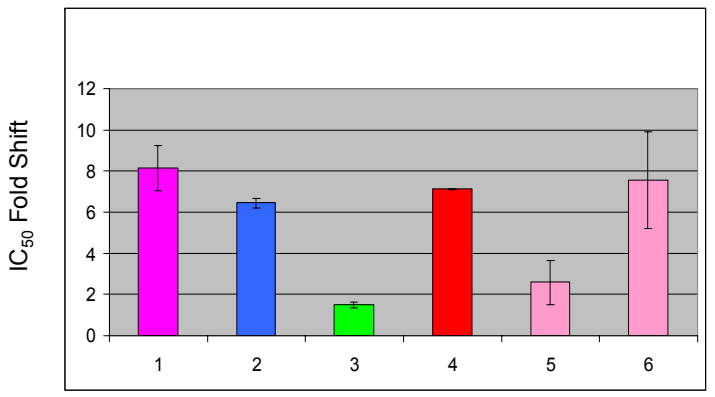
IC50 Fold Shift

metRS1 antisense clones 1-6

- Clones 1-6 were tested against RX-100019 which targets metRS1
- Antisense clones showed differential hypersensitivity to the target compound

Materials and Methods Cont.

Specific Hypersensitivity Testing: Twelve *murA1* antisense clones and six *metRS1* antisense clones were tested in dose response curves to drugs in the presence and absence of 40 mM (*murA1*) or 60 mM (*metRS1*) xylose. The MurA-specific drug phosphomycin was used in testing the *murA1* antisense clones, and the MetRS-specific compound Rx-100019 was used in testing the *metRS1* antisense clones. The IC_{50} shift is defined as the ratio of the 50% growth inhibition concentration for the drug in the absence and presence of xylose.

Results

- Of the twelve *murA1* antisense clones (Figure 1) tested against the MurA-specific drug phosphomycin, only six showed IC_{50} shifts greater than five (Figure 3)
- Similarly, of the six *metRS1* antisense clones (Figure 5) tested with Rx-100019, only four showed IC_{50} shifts greater than five (Figure 6)
- Two *murA1* antisense clones: 1 and 8 showed drastically different profiles against an expanded panel of known antibiotics. Clone 1 showed higher sensitivity to penicillin binding protein inhibitors and other cell wall inhibitors compared to the clone 8 (Figure 4). Both clones showed no IC_{50} shift to drugs targeting protein synthesis, lipid biosynthesis, DNA synthesis, and other cellular functions.

Conclusions

- Antisense clones respond differently to a selective drug target
- Clones showed different IC_{50} shifts to antibacterial compounds targeting the gene product
- Comparison of two *murA1* clones showed another level of differentiation, namely IC_{50} shifts to PBP inhibitors
- For optimal utility as a mode of action detection tool, it is useful to generate a number of antisense clones and select those with a better profile

References

1. Kedar G.C. et al., (2005) 45th ICAAC Washington D.C. F-2075
2. Kedar G.C. et al., *Antimicrobial Agents and Chemotherapy*, 2007. 51: p. 1708-1718
3. Haselbeck et. al., (2006) 46th ICAAC San Franscisco , CA F2-1170
4. Du et al., J Bacteriol, 182:4146-4152
5. Forsyth, R.A., et al., *Mol. Microbiol.* 2002. 43:1387-400
6. Ji, Y., et al., *Science*, 2001. 293:2266-9
7. Gardete, S., et al., *J Bacteriol*, 2004. 186:1705-13
8. Sobral, R.G., et al., *Microb Drug Resist*, 2003. 9:231-41
9. Nakazawa et al., J Infect Chemotherapy, 2003, 9:304-309
10. Real et al., J. Bacteriol, 2006, 188:1721-1732
11. Quinn, C.P., et al., *J Biol Chem*, 1991. 266:20124-30